1353

Effects of NE3107 Anti-Inflammatory Treatment on Motor Activity and Neurodegenerative Features of Parkinson's Disease in a Marmoset Monkey Model

Ingrid HCHM Philippens[1], Clarence Ahlem[2], Christopher L Reading[2]

[1]Charles River Laboratories, Beerse, Belgium; [2]BioVie Inc., Carson City, Nevada, USA.

BACKGROUND

Figure 1. PD pathogenesis

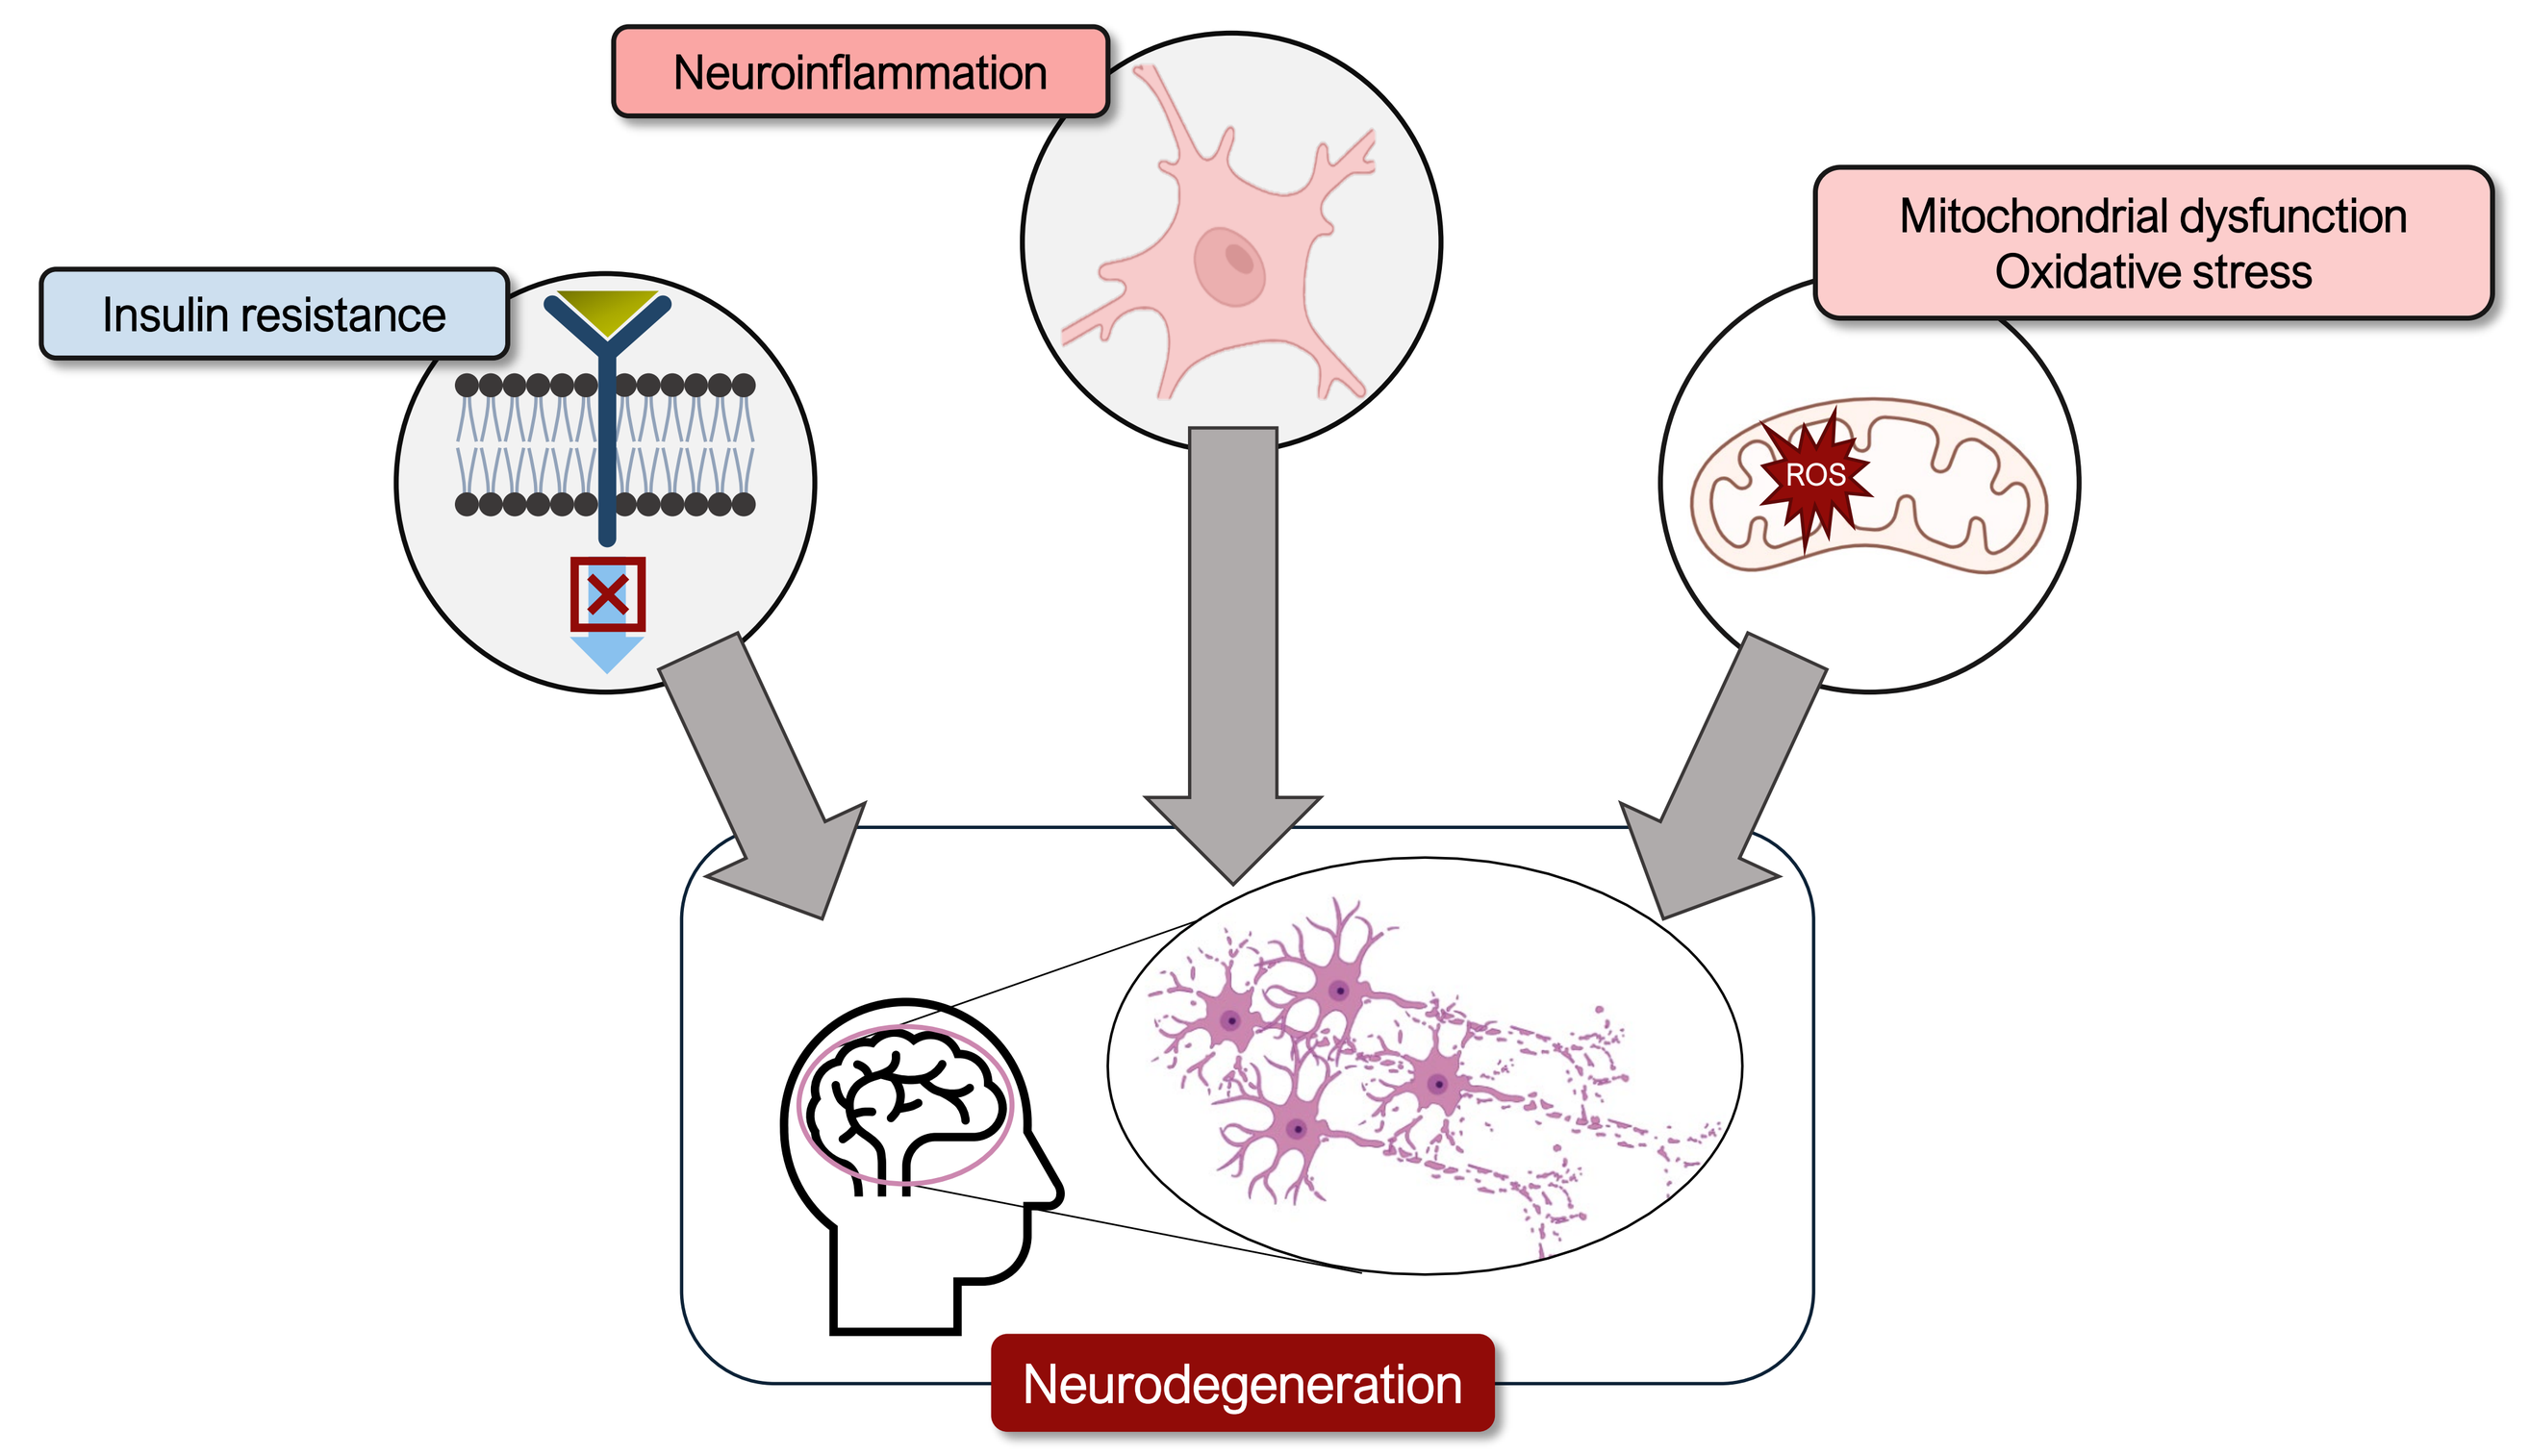

Neuroinflammation
Insulin resistance
Mitochondrial dysfunction
Oxidative stress
ROS
Neurodegeneration

For more information see Athauda D, Foltynie T, 2016[1]; and Dias V, Junn E, Mouradian MM, 2013[2]
ROS, reactive oxygen species.

- Chronic neuroinflammation, aberrant insulin signaling, and oxidative stress are key processes that drive neurodegeneration in Parkinson's disease (PD; **Figure 1**)[1,2]
- Levodopa, a dopamine precursor, is an effective first-line treatment for PD and improves motor function, but it lacks disease-modifying potential, and long-term exposure can lead to diminished efficacy and involuntary movements called levodopa-induced dyskinesia (LID), due to dopamine hypersensitivity[3-5]
 - There is a great need for non-dopaminergic, safe, and effective therapeutic alternatives to levodopa that can slow/prevent the progression of PD
- NE3107 is an oral, blood-brain–permeable, anti-inflammatory, and insulin-sensitizing molecule that binds to the inflammatory mediator ERK and inhibits inflammation-specific ERK and NF-κB activation and TNF-α signaling, without affecting homeostasis[6]
 - In the context of PD, NE3107 may downregulate microglia to reduce purine- and glutamate-stimulated inhibition of dopamine receptors, thereby improving motor function[7]
- NE3107 has been shown to have a favorable safety profile and demonstrated anti-inflammatory, insulin-sensitizing, pro-motoric, and neuroprotective effects in pre-clinical studies using rodents[4,6,8,9]
- To bolster the rationale for investigating NE3107 in human PD clinical trials, we used an MPTP-induced marmoset model of PD and studied the potential effects of NE3107 on disease pathology and motor function

OBJECTIVES

- The overall objectives of this pre-clinical study were to evaluate the safety, tolerability, and the extent of the anti-inflammatory effects of NE3107 treatment, specifically on the major features of PD, in marmosets with Parkinson's-like disease

METHODS

Study Design

- In this 14-week study, Parkinson's-like disease and LID were induced in marmosets (*Cllithrix jacchus*) aged 2-5 years, and the effects of NE3107 (30 mg/kg daily), amantadine HCl (1 mg/kg daily), or vehicle (placebo; acacia syrup) were evaluated (**Figure 2**)
 - This study was conducted in a series of three sequential independent cohorts of 6 marmosets
 - Week 4: single dose of NE3107 or amantadine HCl to ensure the absence of behavioral side effects
 - Weeks 5-6: subcutaneous injections of 1-2 mg/kg MPTP on days 3-5 of week 5 and days 1 and 3 of week 6
 - Week 7: at the end of week 7, animals were randomized to receive NE3107, amantadine HCl, or control
 - Weeks 8-9: treatment effects against parkinsonian behavior were evaluated
 - Weeks 10-11: escalating doses of levodopa (5, 7.5, 10, and 12.5 mg/kg) were administered along with treatment, and effects on parkinsonian behavior were evaluated
 - Weeks 12-13: LID was induced in all animals with 12.5 mg/kg levodopa BID along with treatment, and the severity of LID was measured using Abnormal Involuntary Movement Score (AIMS)
 - Week 14: treatment was administered without levodopa, and animals were euthanized for brain immunohistochemical analyses

Figure 2. Study design

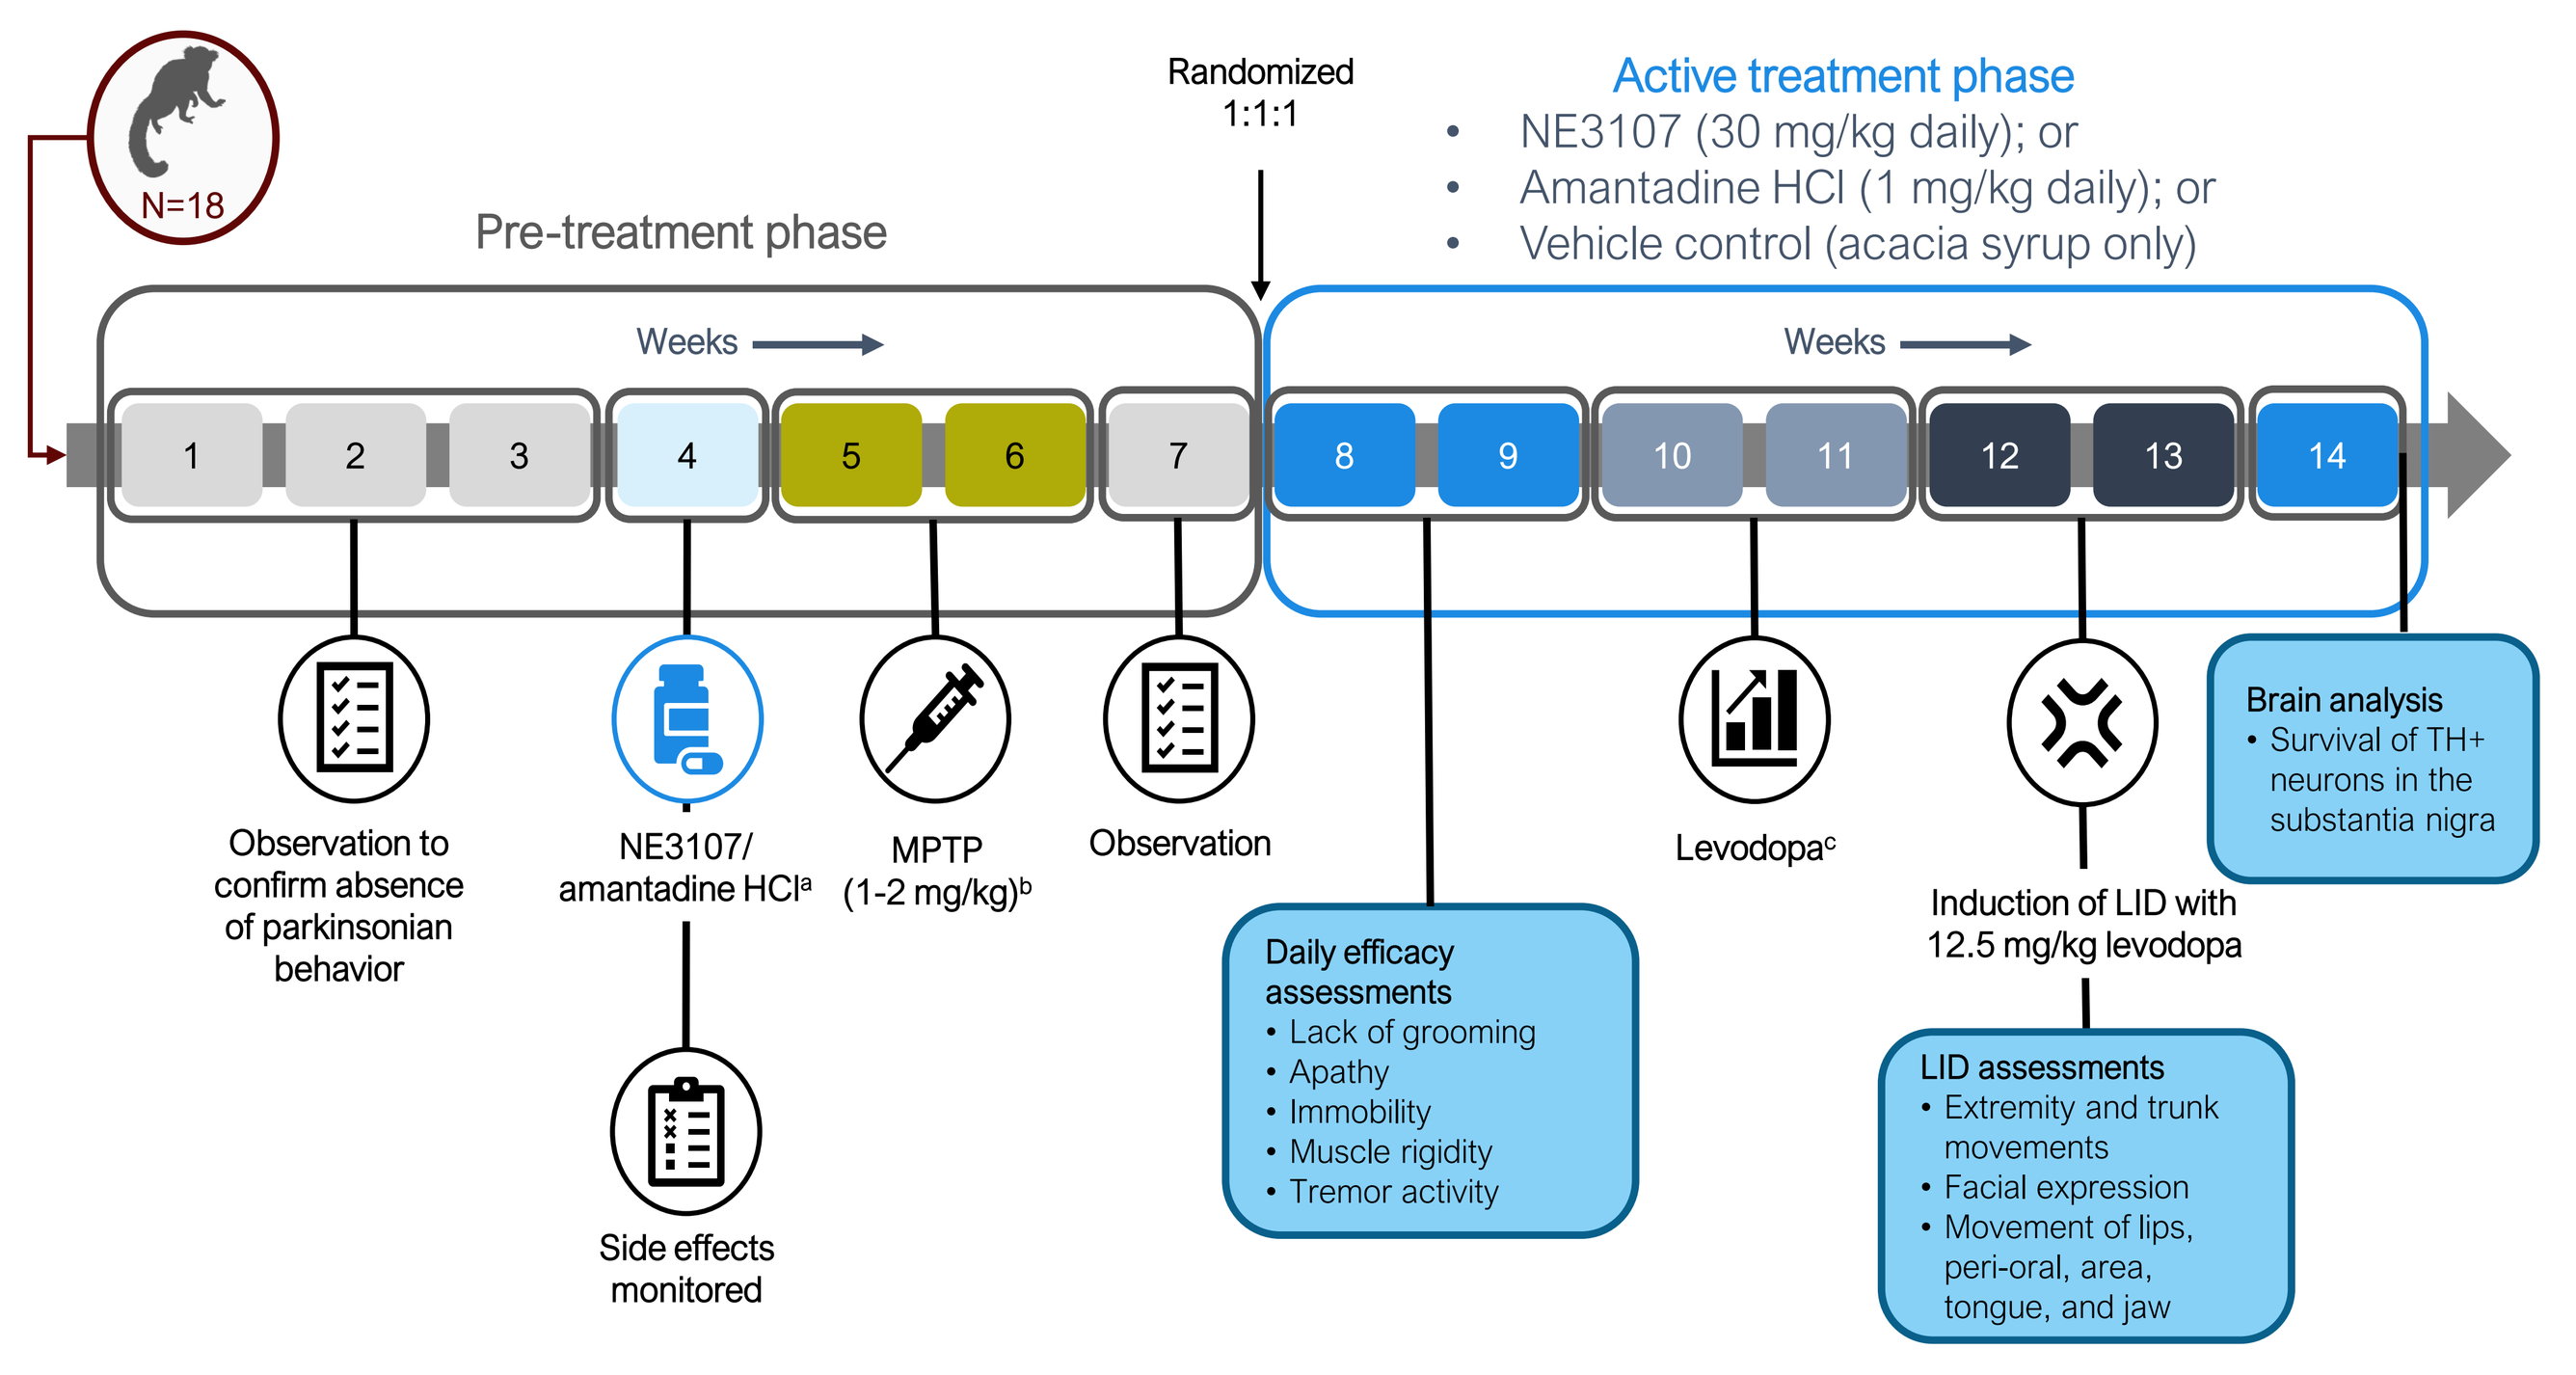

N=18
Pre-treatment phase
Randomized 1:1:1
Active treatment phase
• NE3107 (30 mg/kg daily); or
• Amantadine HCl (1 mg/kg daily); or
• Vehicle control (acacia syrup only)
Weeks
1 2 3 4 5 6 7 8 9 10 11 12 13 14
Observation to confirm absence of parkinsonian behavior
NE3107/ amantadine HCl[a]
MPTP (1-2 mg/kg)[b]
Observation
Side effects monitored
Daily efficacy assessments
• Lack of grooming
• Apathy
• Immobility
• Muscle rigidity
• Tremor activity
Levodopa[c]
Induction of LID with 12.5 mg/kg levodopa
LID assessments
• Extremity and trunk movements
• Facial expression
• Movement of lips, peri-oral, area, tongue, and jaw
Brain analysis
• Survival of TH+ neurons in the substantia nigra

[a]Single dose. [b]Administered subcutaneously on days 3-5 of week 5 and days 1 and 3 of week 6 for a total of 6.5 mg/kg. [c]Single escalating doses of 5, 7.5, 10, and 12.5 mg/kg.
LID, levodopa-induced dyskinesia; MPTP, 1-methyl-4-phenyl-1,2,3,6-tetrahydropyridine; TH, tyrosine hydroxylase.

Assessments

Treatment effects on parkinsonian behavior – weeks 8-11; scored on a 0 (normal) – 4 (most severe) scale

- Lack of grooming
- Apathy
- Immobility
- Muscle rigidity
- Tremor activity

Severity of LID – weeks 12-13; measured by AIMS (adapted to the primate model)

- Assessment of:
 - Extremity and trunk movements
 - Facial expression
 - Movement of the lips, peri-oral area, tongue, and jaw
- Rating scale: 0 (normal), 1 (extreme normal), 2 (mild), 3 (moderate) to 4 (severe)

Brain immunohistochemical analysis – end of week 14

- DA-positive neurons within the SNpc using TH-IR staining

RESULTS

Tolerability

- NE3107 treatment was well tolerated
- One monkey randomized to amantadine HCl and one randomized to NE3107 died prior to treatment

Immobility analysis

- Mean immobility scores with NE3107 monotherapy during weeks 8 and 9 were significantly lower (indicating improved mobility) compared with amantadine HCl treatment or vehicle only (**Table 1**)
- Immobility scores significantly improved with NE3107 in combination with levodopa doses ≥7.5 mg/kg/day during weeks 10-11, compared with NE3107 monotherapy (**Figure 3**)

Table 1. Mean immobility scores with monotherapy

Week	Vehicle	Amantadine HCl	NE3107	*P* (vehicle vs amantadine)	*P* (NE3107 vs vehicle)	*P* (NE3107 vs amantadine)
7[a]	2.71	2.59	2.72	0.58	0.86	0.433
8	3.0	2.74	2.26	0.104	<0.0001	0.0048
9	2.8	2.72	2.10	0.75	0.00088	0.0044

[a]Represents scores at the end of disease induction and prior to initiation of therapy.

Effect of treatment on LID (evaluated using AIMS)

- During week 12, all animals developed LID following administration of 12.5 mg/kg levodopa BID
- NE3107 + levodopa combination treatment was associated with significantly lower AIMS compared with vehicle + levodopa treatment (mean: 2.1 vs 7.2; *P*<0.0001) and amantadine HCl + levodopa treatment (mean: 2.1 vs 5.4; *P*<0.0001) (**Table 2**)
 - NE3107 + levodopa combination treatment was associated with fewer LID observations compared with either vehicle or amantadine + levodopa combination treatments (**Figure 4**)

Figure 3. Mean immobility scores (weeks 10 and 11) with combination treatment

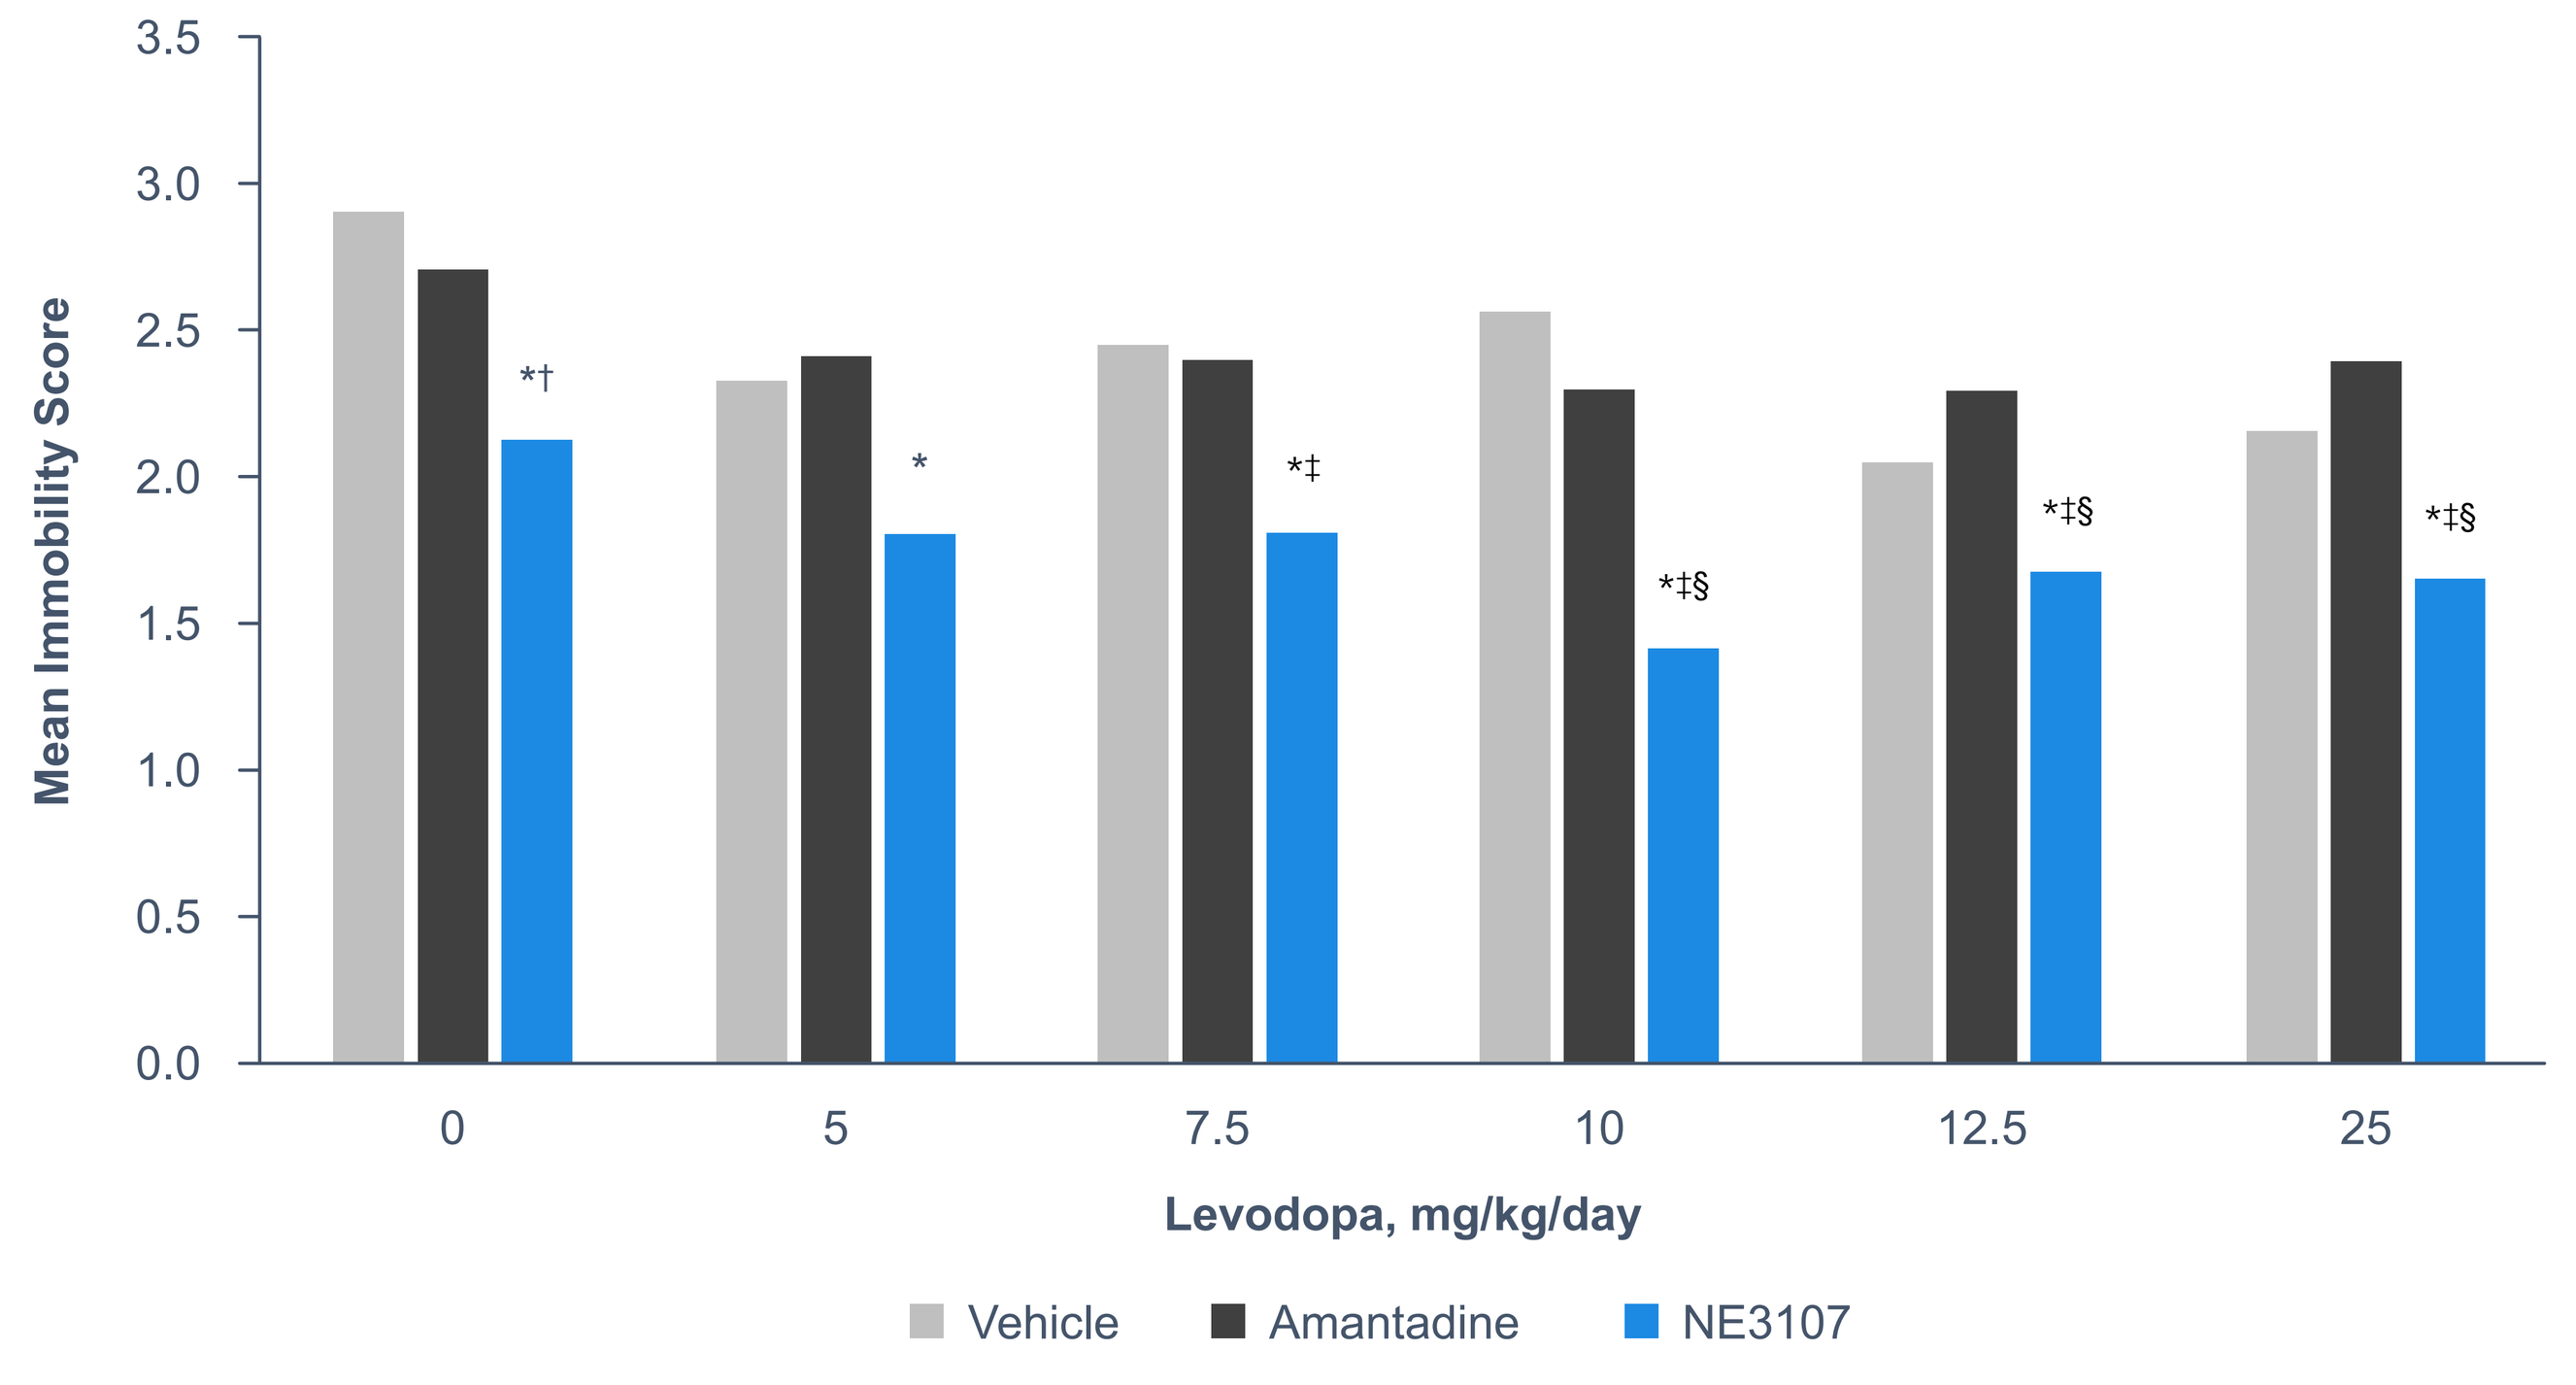

Mean Immobility Score
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
*†
*
*‡
*‡§
*‡§
*‡§
0
5
7.5
10
12.5
25
Levodopa, mg/kg/day
Vehicle
Amantadine
NE3107

*P<0.05 vs vehicle and vehicle + levodopa; †P<0.05 vs amantadine HCl monotherapy; ‡P<0.05 vs NE3107 monotherapy; §P<0.05 vs amantadine HCl + levodopa. L-dopa, levodopa.

Table 2. Mean AIMS for week 12

	Vehicle	Amantadine HCl	NE3107
AIMS	7.2	5.4	2.1
P[a] (vs vehicle)	-	0.094	<0.0001
P[a] (vs amantadine HCl)	0.094	-	<0.0001

[a]Student's t-Test (two-sided assuming equal variance). AIMS, Abnormal Involuntary Movement Score.

Figure 4. Effect of combination treatment on LID (AIMS)

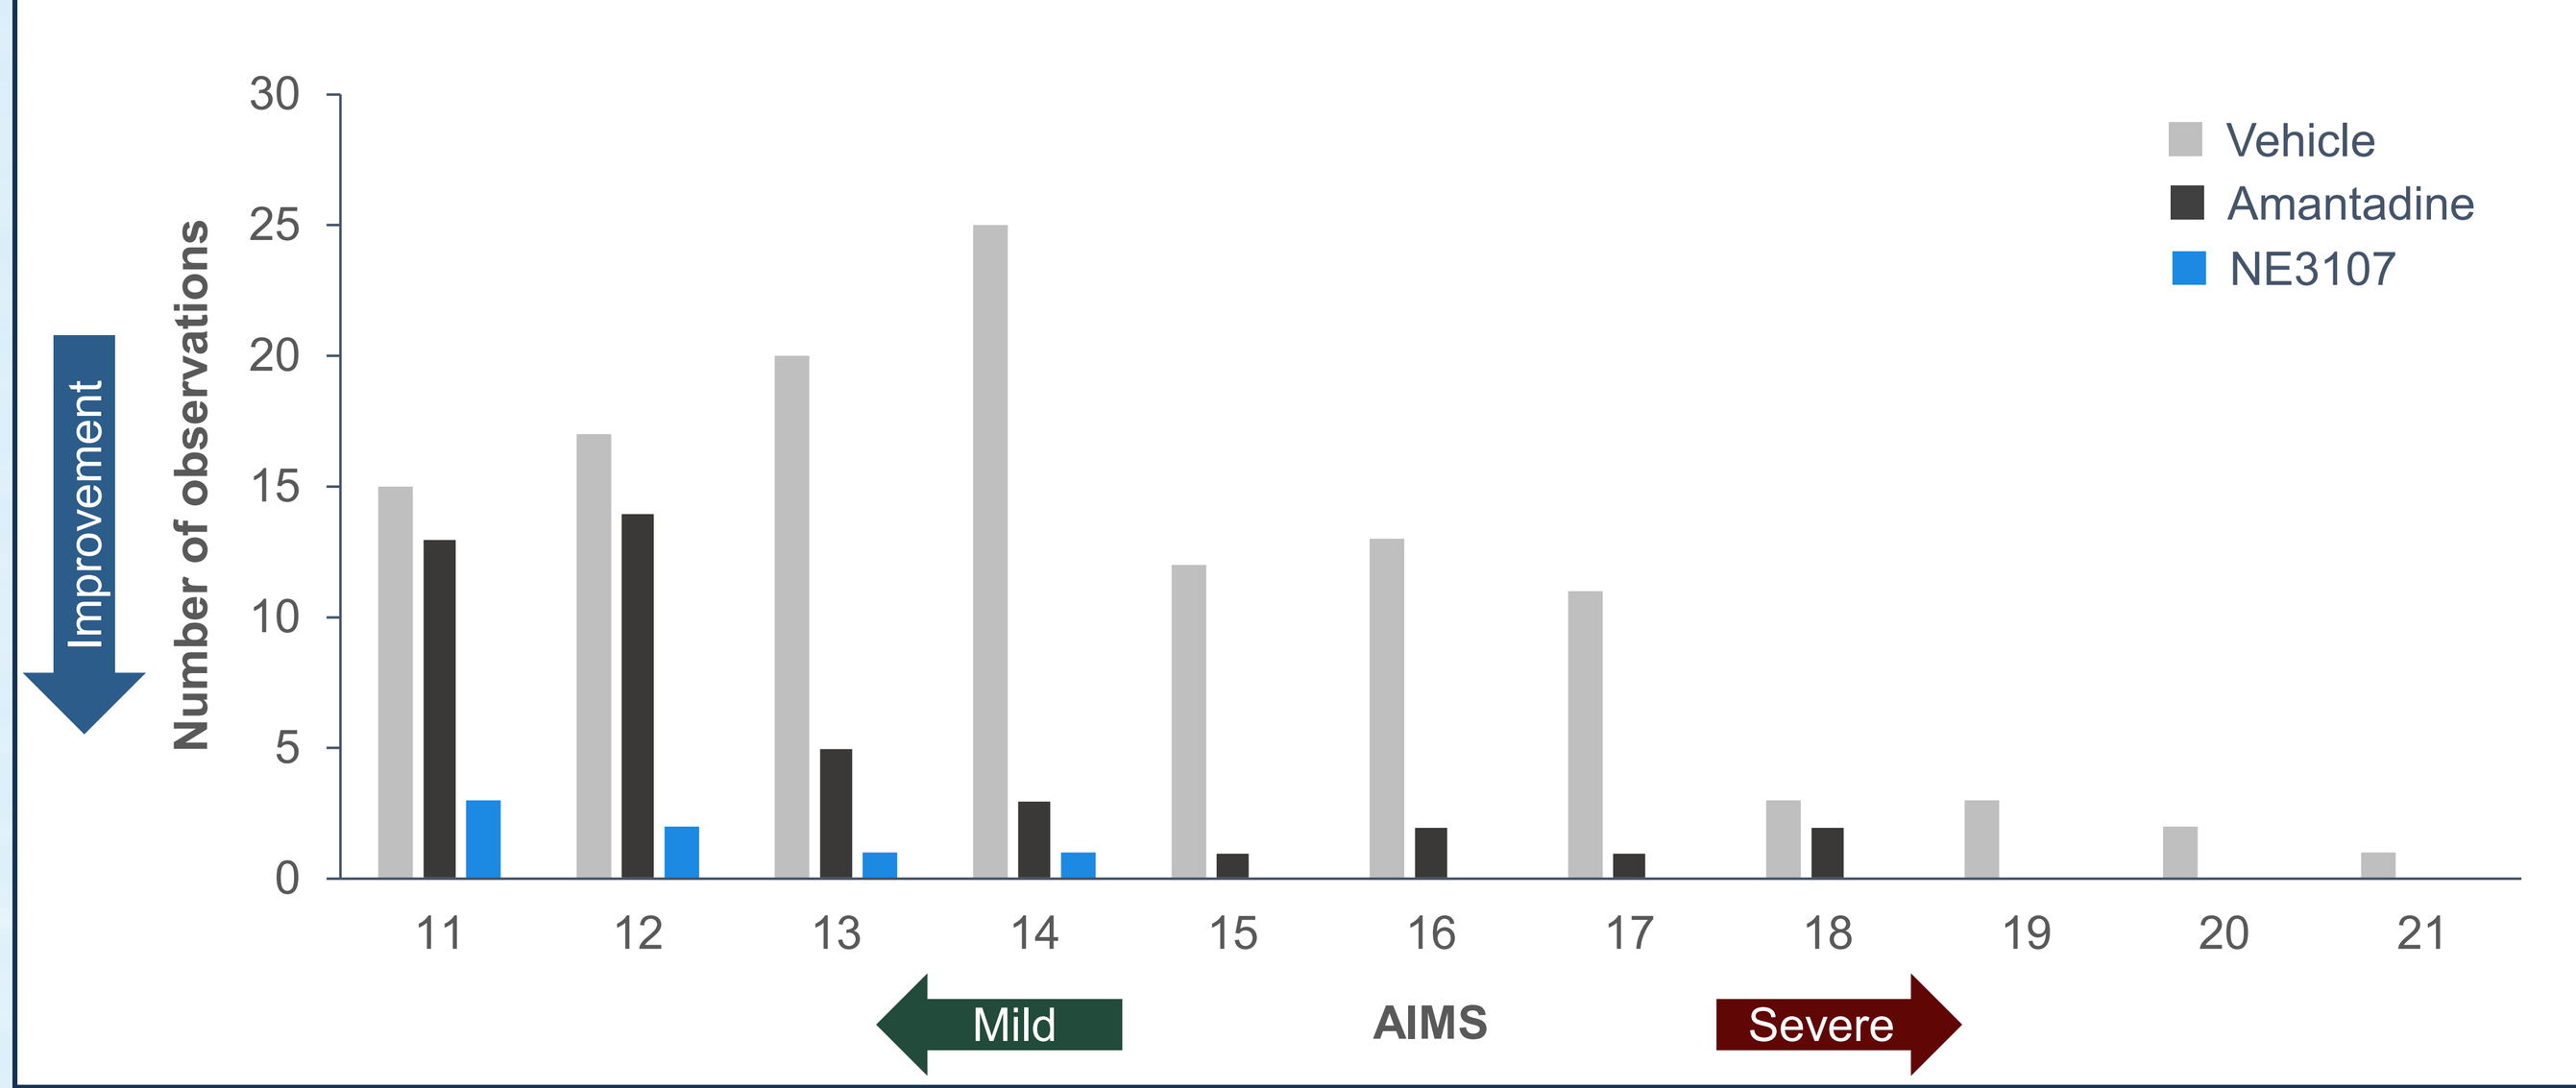

Improvement
Number of observations
30
25
20
15
10
5
0
11
12
13
14
15
16
17
18
19
20
21
Mild
AIMS
Severe
Vehicle
Amantadine
NE3107

Brain immunohistochemical analysis

- Monkeys treated with NE3107, but not amantadine HCl, had significantly more surviving TH+ neurons compared with those treated with vehicle control (*P*=0.0108; **Figures 5** and **6**)

Figure 5. Relative TH+ neuron survival in the substantia nigra

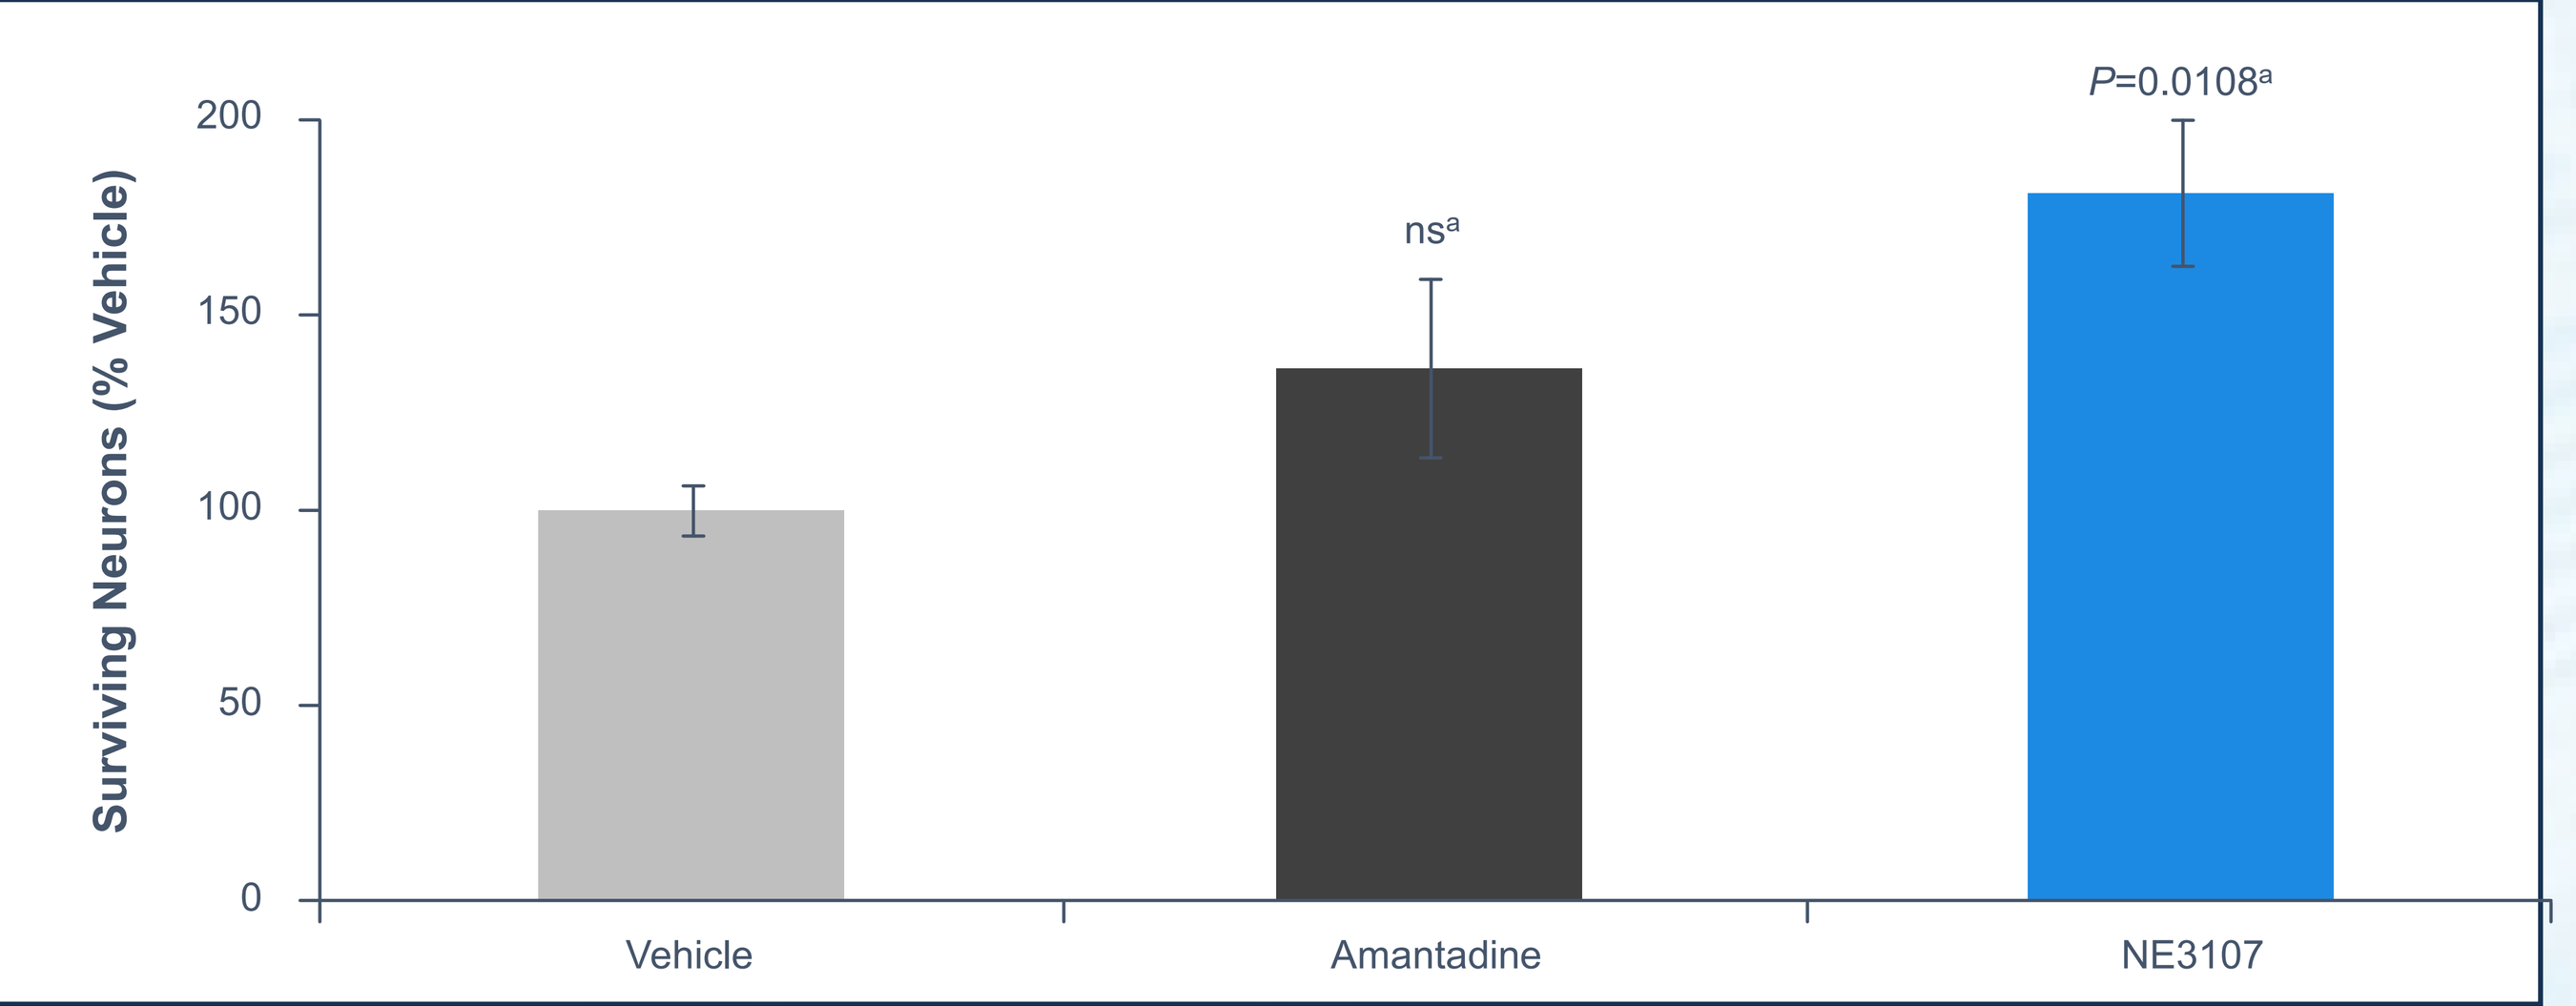

Surviving Neurons (% Vehicle)
200
150
100
50
0
ns[a]
P=0.0108[a]
Vehicle
Amantadine
NE3107

[a]vs vehicle. ns, non-significant; TH, tyrosine hydroxylase.

Figure 6. Representative brain samples showing TH+ cells (TH-IR staining)

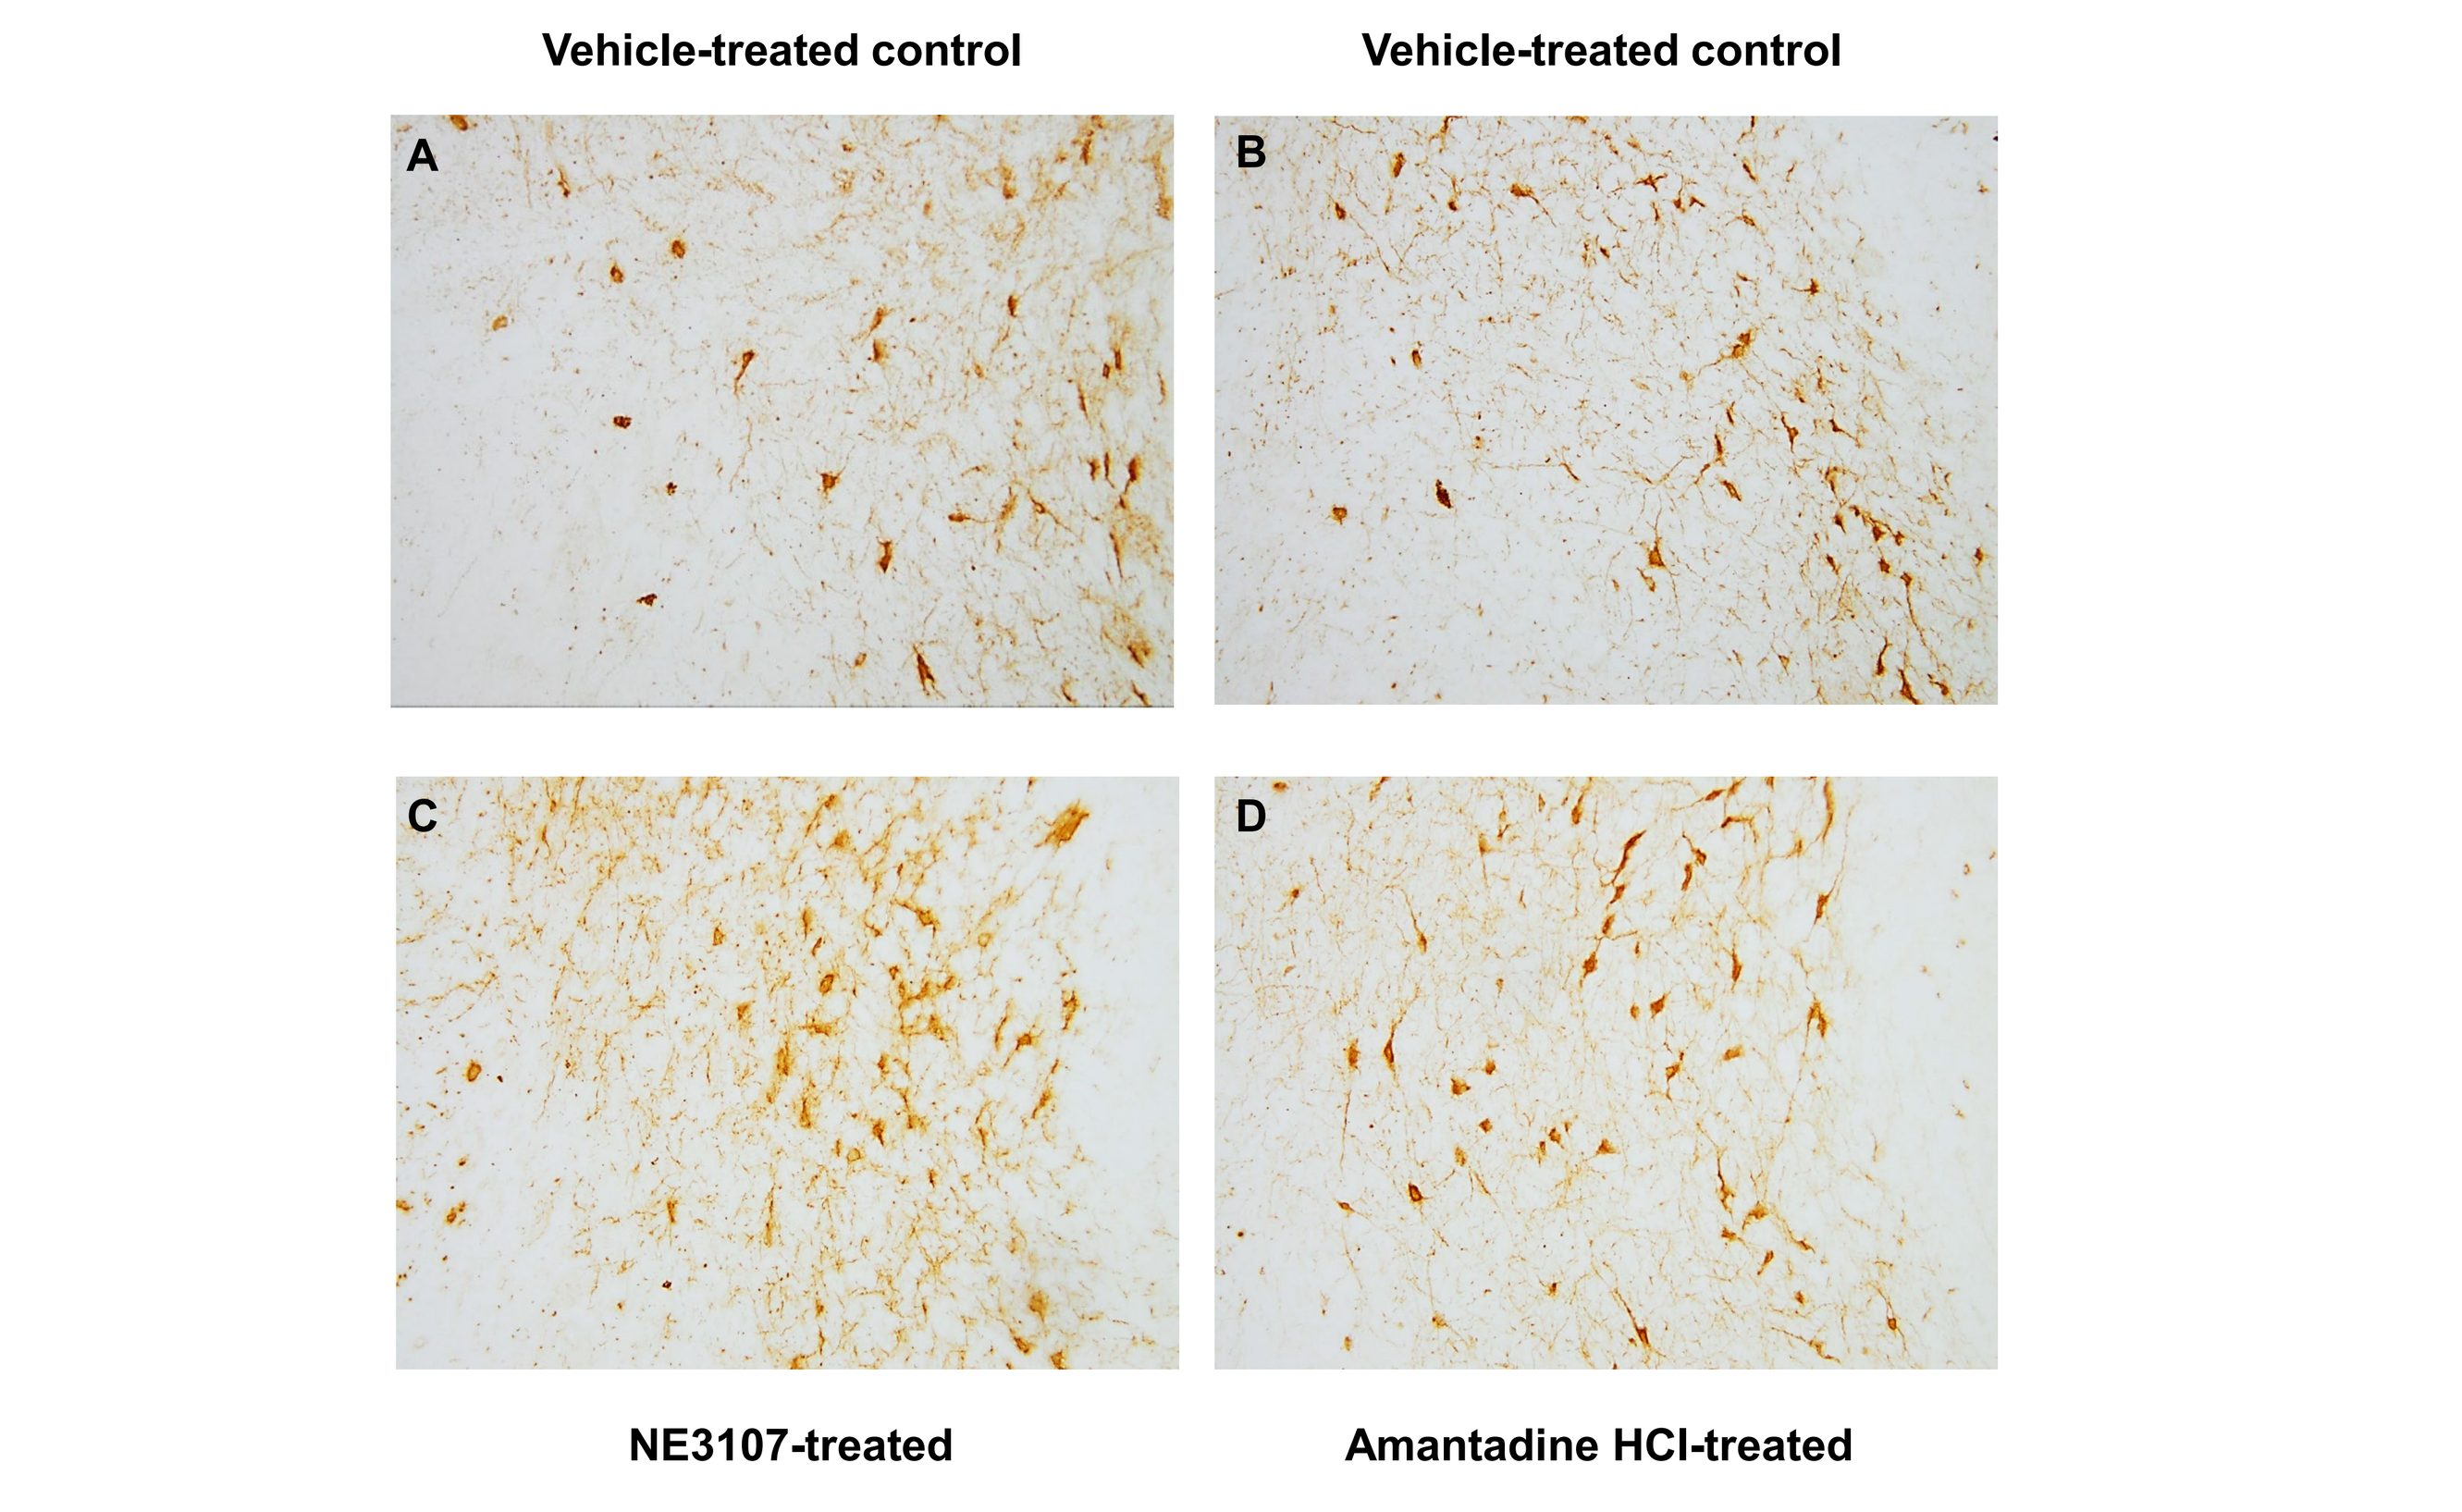

Vehicle-treated control
Vehicle-treated control
A
B
C
D
NE3107-treated
Amantadine HCl-treated

TH, tyrosine hydroxylase; TH-IR, tyrosine hydroxylase–immune reactive.

CONCLUSIONS

- In this 14-week pre-clinical trial of 18 marmoset monkeys with MPTP-induced Parkinson's-like disease, NE3107 monotherapy was associated with significantly lower immobility scores than treatment with either amantadine HCl or vehicle alone
- Improvements in mobility were observed within 24 hours of initiating NE3107 monotherapy, suggesting a direct influence on neuro-motor signaling, as opposed to an effect that increased with time of treatment, which might imply a link to neuroprotection
- NE3107 was significantly more effective in alleviating symptoms of LID in monkeys, compared with treatment with either amantadine HCl or vehicle
 - Importantly, the reduction in LID was achieved simultaneously with improvements in mobility
 - Key mediators of neuroinflammation such as ERK, NF-κB, and TNF-α have been implicated in the pathogenesis of LID[10-12]
 - The risk of LID represents an important unmet medical need in PD as patients are often unable to benefit fully from levodopa due to its potential to cause dyskinesia[5]
- Monkeys treated with NE3107, but not amantadine HCl, had significantly more surviving TH+ neurons in the SNpc, indicating decreased neurodegeneration
 - TH is an essential enzyme in the biosynthesis of dopamine, and its deficiency is integral to PD progression[13]
 - Neuroinflammation and aberrant insulin signaling are thought to contribute to neurodegeneration in PD, and inhibition of key neuroinflammatory mediators may have neuroprotective effects[1]
- Our findings corroborate the involvement of chronic neuroinflammation and insulin resistance in PD clinical symptoms, in addition to neurodegenerative pathways, and support the continued investigation of nondopaminergic therapies
- A recently completed phase 2, double-blind, placebo-controlled trial (NCT05083260) evaluated the safety and efficacy of NE3107 in 45 levodopa-treated patients with PD

REFERENCES

1. Athauda D, Foltynie T. Insulin resistance and Parkinson's disease: a new target for disease modification? *Prog Neurobiol*. 2016;145-146:98-120.
2. Dias V, Junn E, Mouradian MM. The role of oxidative stress in Parkinson's disease. *J Parkinsons Dis*. 2013;3(4):461-491.
3. National Institute of Neurological Disorders and Stroke. *Parkinson's disease: challenges, progress, and promise*. NIH/NINDS website. Accessed July 20, 2023. https://www.ninds.nih.gov/current-research/focus-disorders/focus-parkinsons-disease-research/parkinsons-disease-challenges-progress-and-promise
4. Nicoletti F, Philippens I, Fagone P, et al. 17α-Ethynyl-androst-5-ene-3β,7β,17β-triol (HE3286) is neuroprotective and reduces motor impairment and neuroinflammation in a murine MPTP model of Parkinson's disease. *Parkinsons Dis*. 2012;2012:969418. doi:10.1155/2012/969418
5. LeWitt PA, Chaudhuri KR. Unmet needs in Parkinson disease: motor and non-motor. *Parkinsonism Relat Disord*. 2020;80(suppl 1):S7-S12.
6. Reading CL, Ahlem CN, Murphy MF. NM101 phase III study of NE3107 in Alzheimer's disease: rationale, design and therapeutic modulation of neuroinflammation and insulin resistance. *Neurodegener Dis Manag*. 2021;11(4):289-298.
7. Beggiato S, Tomasini MC, Borelli AC, et al. Functional role of striatal A2A, D2, and mGlu5 receptor interactions in regulating striatopallidal GABA neuronal transmission. *J Neurochem*. 2016;138(2):254-264.
8. Khan RS, Dine K, Luna E, Ahlem C, Shindler KS. HE3286 reduces axonal loss and preserves retinal ganglion cell function in experimental optic neuritis. *Invest Ophthalmol Vis Sci*. 2014;55(9):5744-5751.
9. Lambert WS, Carlson BJ, Formichella CR, Sappington RM, Ahlem C, Calkins DJ. Oral delivery of a synthetic sterol reduces axonopathy and inflammation in a rodent model of glaucoma. *Front Neurosci*. 2017;11:45. doi:10.3389/fnins.2017.00045
10. Dos Santos Pereira M, Abreu GHD, Rocca J, et al. Contributive role of TNF-α to L-DOPA-induced dyskinesia in a unilateral 6-OHDA lesion model of Parkinson's disease. *Front Pharmacol*. 2021;11:617085. doi:10.3389/fphar.2020.617085
11. Feyder M, Bonito-Oliva A, Fisone G. L-DOPA-induced dyskinesia and abnormal signaling in striatal medium spiny neurons: focus on dopamine D1 receptor-mediated transmission. *Front Behav Neurosci*. 2011;5:71. doi:10.3389/fnbeh.2011.00071
12. Wan Y, Han L, Rong L, et al. Inhibition of BET protein function suppressed the overactivation of the canonical NF-κB signaling pathway in 6-OHDA-lesioned rat model of levodopa-induced dyskinesia. *Front Neurosci*. 2022;16:896322. doi:10.3389/fnins.2022.896322
13. Tabrez S, Jabir NR, Shakil S, et al. A synopsis on the role of tyrosine hydroxylase in Parkinson's disease. *CNS Neurol Disord Drug Targets*. 2012;11(4):395-409.

ACKNOWLEDGEMENTS

p-value communications provided editorial support for this presentation, supported by BioVie Inc.

This research was sponsored by a Therapeutics Development Initiative grant from the
Michael J. Fox Foundation for Parkinson's Research

DISCLOSURES

CA and **CLR** are employees of BioVie Inc. **IHCHMP** has nothing to disclose.

BID, twice per day; DA, dopamine; ERK, extracellular signal-regulated kinase; MPTP, 1-methyl-4-phenyl-1,2,3,6-tetrahydropyridine; NF-κB, nuclear factor kappa-light-chain-enhancer of activated B cells; ns, non-significant; SNpc, substantia nigra pars compacta; TH, tyrosine hydroxylase; TH-IR, tyrosine hydroxylase immune reactive; TNF-α, tumor necrosis factor alpha.

Presented at the 2023 International Congress of Parkinson's Disease and Movement Disorders | August 27-31, 2023 | Copenhagen, Denmark